Board 071; Abstract 120

DCC-2618, a broad-spectrum inhibitor of KIT and PDGFRA mutants, synergizes with inhibitors of the MAPK pathway

Anu Gupta[1], Cynthia B. Leary[1], Alfonso Garcia-Valverde[2], Joaquin Arribus[2], Cesar Serrano[2], Daniel L. Flynn[1], Bryan D. Smith[1]

[1]Deciphera Pharmaceuticals, LLC, Waltham, MA; [2]Preclinical Research program, Vall d'Hebron Institute of Oncology (VHIO), Vall d'Hebron University Hospital, Barcelona, Spain

EORTC-NCI-AACR Annual Meeting, Nov 13-16, 2018, Dublin, Ireland

Introduction

Treatment of gastrointestinal stromal tumors (GIST) with the KIT-inhibitor imatinib leads to objective responses or stable disease, but complete responses are rare. Residual signaling through the MAPK pathway via incomplete suppression of KIT, or activation of other pathways that feed into MAPK signaling are possible mechanisms allowing GIST cells to escape death. The stasis of GIST tumors in response to treatment with KIT inhibitors can lead to rebound tumor growth in the absence of drug or allow for drug-resistance mutations to arise. Inhibitors of the MAPK signaling pathway, such as MEK inhibitors, have been shown to synergize with imatinib to inhibit GIST cell growth *in vitro* and *in vivo (Ran, Cancer Disc. 2015).* DCC-2618 is a kinase switch control inhibitor that broadly and potently inhibits the spectrum of primary and drug-resistant mutations in KIT spanning six exons (9, 11, 13, 14, 17 and 18) and in PDGFRA spanning three exons (12, 14 and 18) that occur in GIST and other diseases. We hypothesized that broadly inhibiting clinically-relevant primary and drug-resistant KIT mutations with DCC-2618 would synergize with MEK inhibitors *in vitro* and *in vivo* in both imatinib-sensitive and imatinib-resistant cell lines and tumors.

Simplified Model of KIT and MAPK Signaling[1]

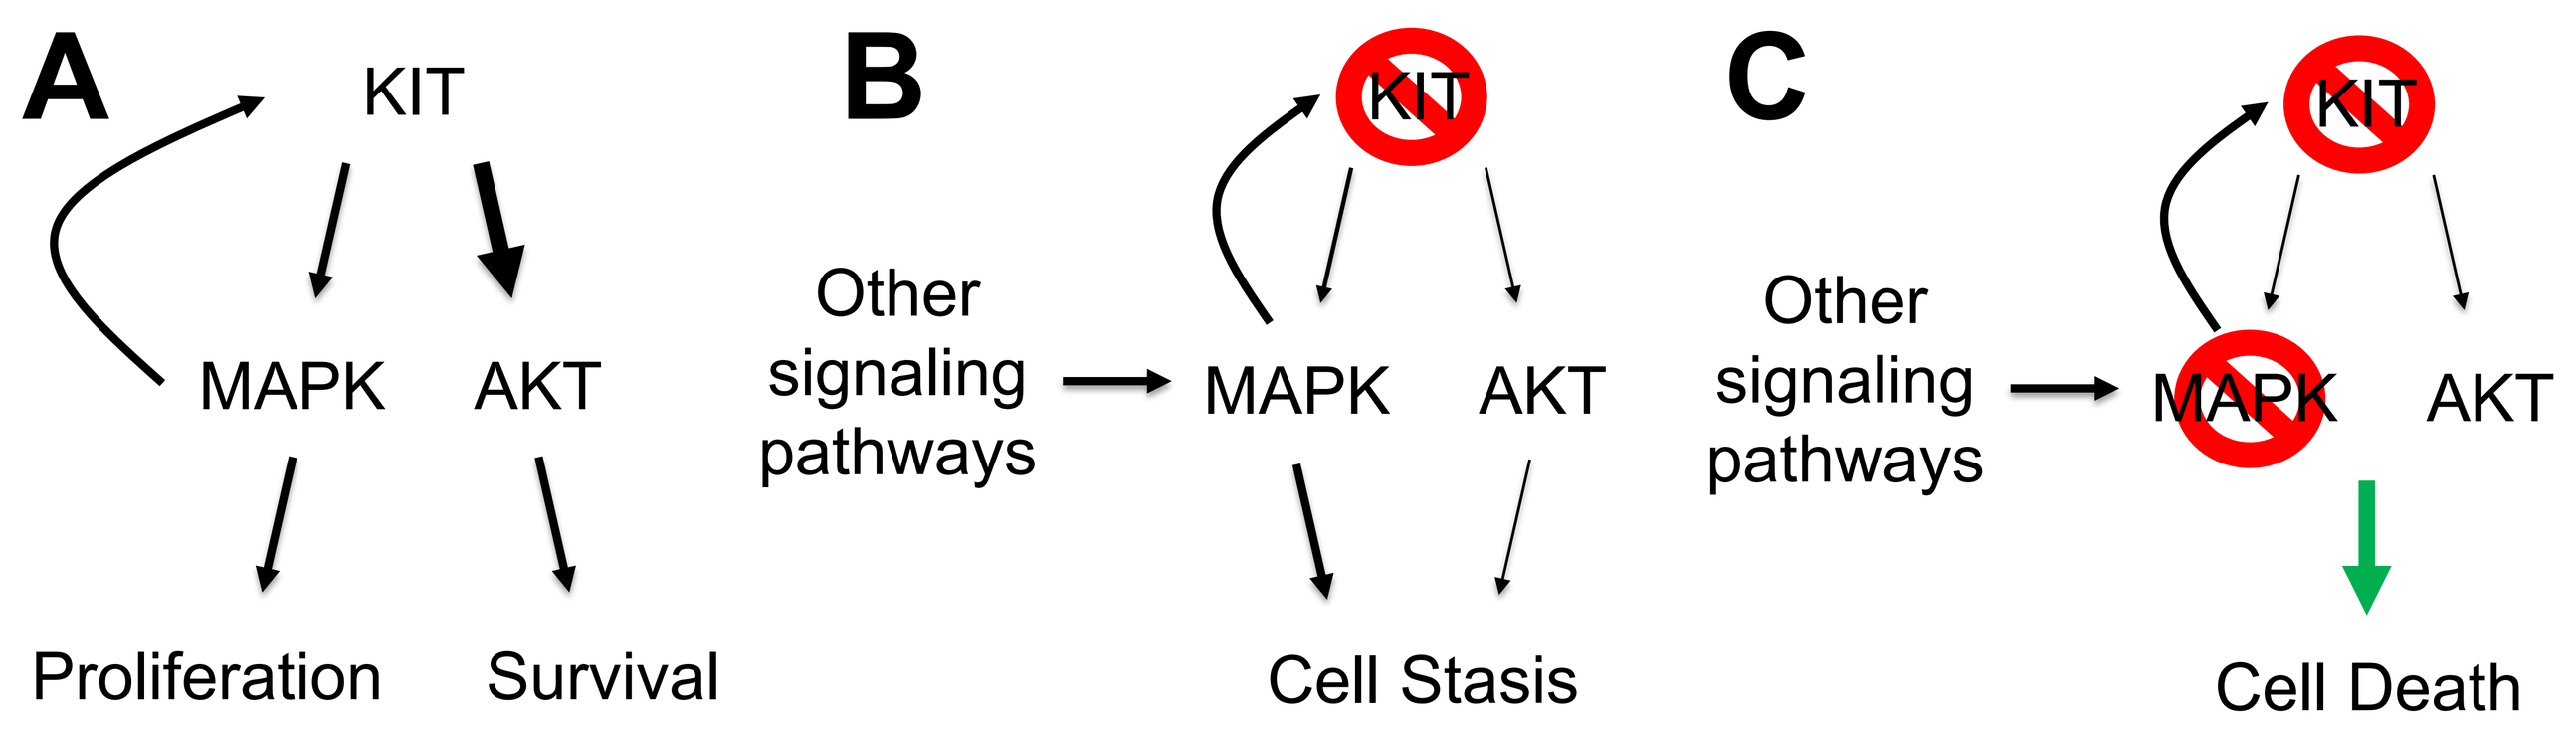
A
KIT
MAPK
AKT
Proliferation
Survival
B
Other signaling pathways
KIT
MAPK
AKT
Cell Stasis
C
Other signaling pathways
KIT
MAPK
AKT
Cell Death

Figure 1: (A) KIT signals through the MAPK and AKT pathways leading to cell proliferation and survival. MAPK activity leads to greater KIT expression. **(B)** KIT inhibition leads to a reduction in MAPK and AKT signaling, but residual MAPK signaling due to feedback or signaling from other pathways can allow cells to escape cell death. **(C)** Combined inhibition of the KIT and MAPK pathways leads to cell death.

[1]Ran L. *et al.* (2015) Combined inhibition of MAP Kinase and KIT Signaling Synergistically Destabilizes ETV1 and Suppresses GIST Tumor Growth. *Cancer Disc.* **5:** 304-315

Combined inhibition of MAPK and KIT signaling induces pro-apoptotic proteins and apoptosis

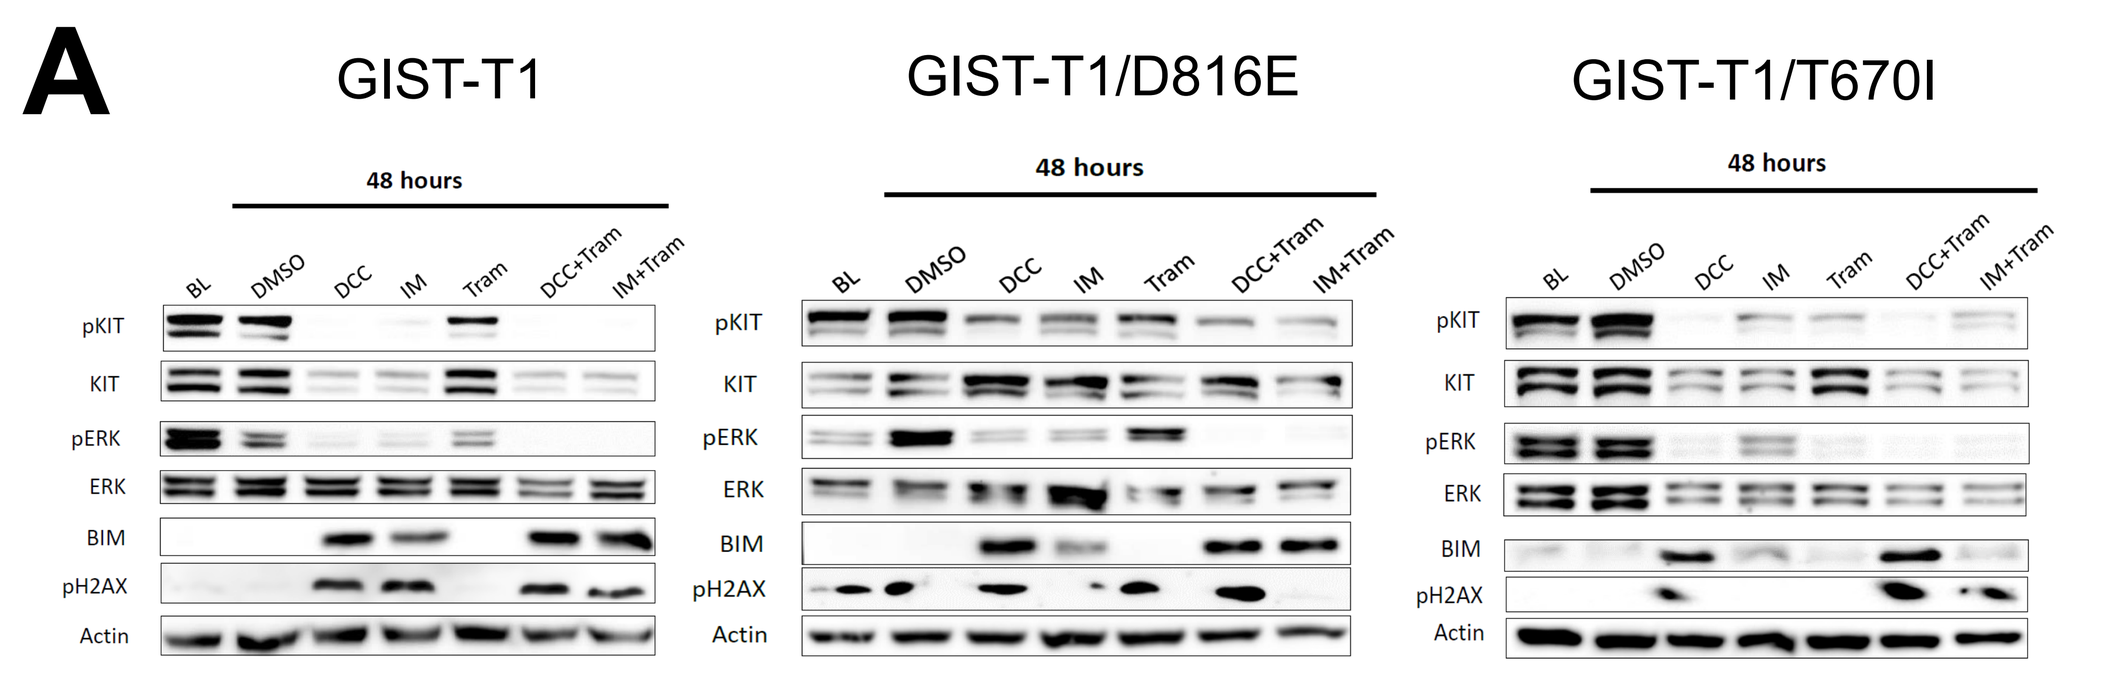
A
GIST-T1
GIST-T1/D816E
GIST-T1/T670I
48 hours
BL
DMSO
DCC
IM
Tram
DCC+Tram
IM+Tram
pKIT
KIT
pERK
ERK
BIM
pH2AX
Actin

Drug	Conc.
imatinib (IM)	100 nM
DCC-2618 (DCC)	100 nM
trametinib (Tram)	10 nM

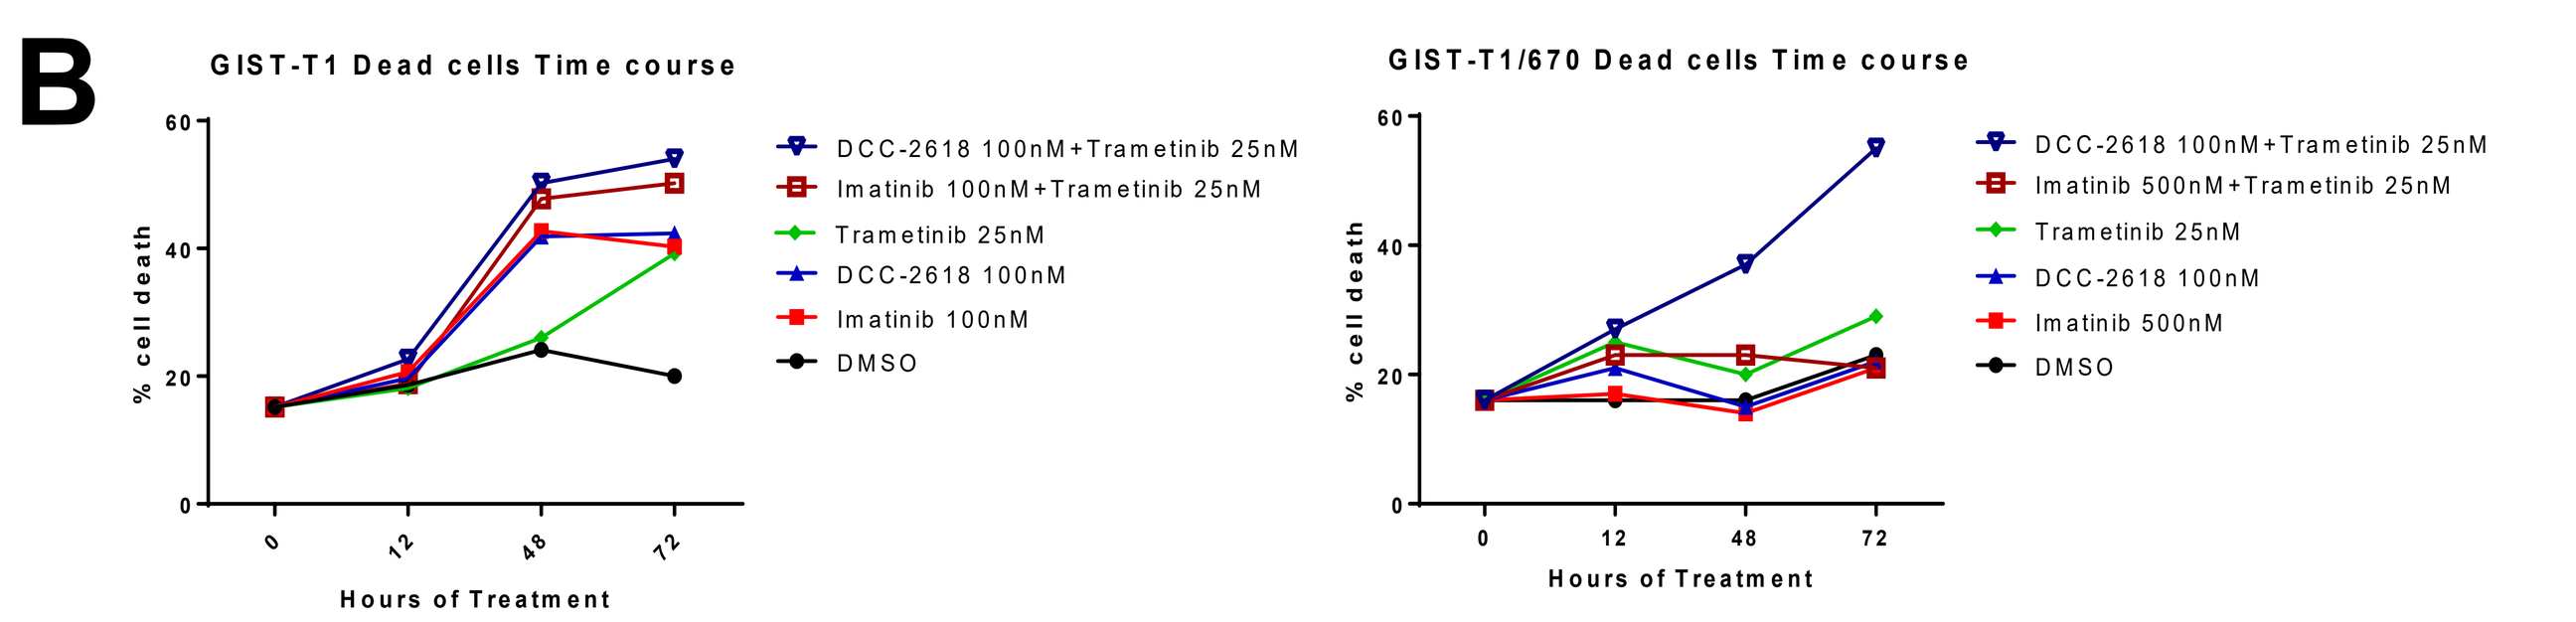
B
GIST-T1 Dead cells Time course
% cell death
Hours of Treatment
DCC-2618 100nM+Trametinib 25nM
Imatinib 100nM+Trametinib 25nM
Trametinib 25nM
DCC-2618 100nM
Imatinib 100nM
DMSO
GIST-T1/670 Dead cells Time course
DCC-2618 100nM+Trametinib 25nM
Imatinib 500nM+Trametinib 25nM
Trametinib 25nM
DCC-2618 100nM
Imatinib 500nM
DMSO

Figure 2: DCC-2618 and trametinib combinations induce apoptosis in GIST cell lines. **(A)** Immunoblot of pKIT, pERK, BIM and pH2AX levels in imatinib-sensitive (GIST-T1) and imatinib-resistant (GIST-T1/D816E and GIST-T1/T670I) cells treated with imatinib (IM, 100 nM) or DCC-2618 (DCC, 100 nM) in the absence or presence of trametinib (Tram, 10 nM) for 48 hr. **(B)** Apoptosis induction in GIST-T1 and GIST-T1-T670I cells as measured by AnnexinV/PI staining. Cells were treated with compounds at indicated concentrations for 24, 48 and 72h and stained with Annexin V and PI.

DCC-2618 synergizes with MEK inhibitors to induce apoptosis in GIST cells

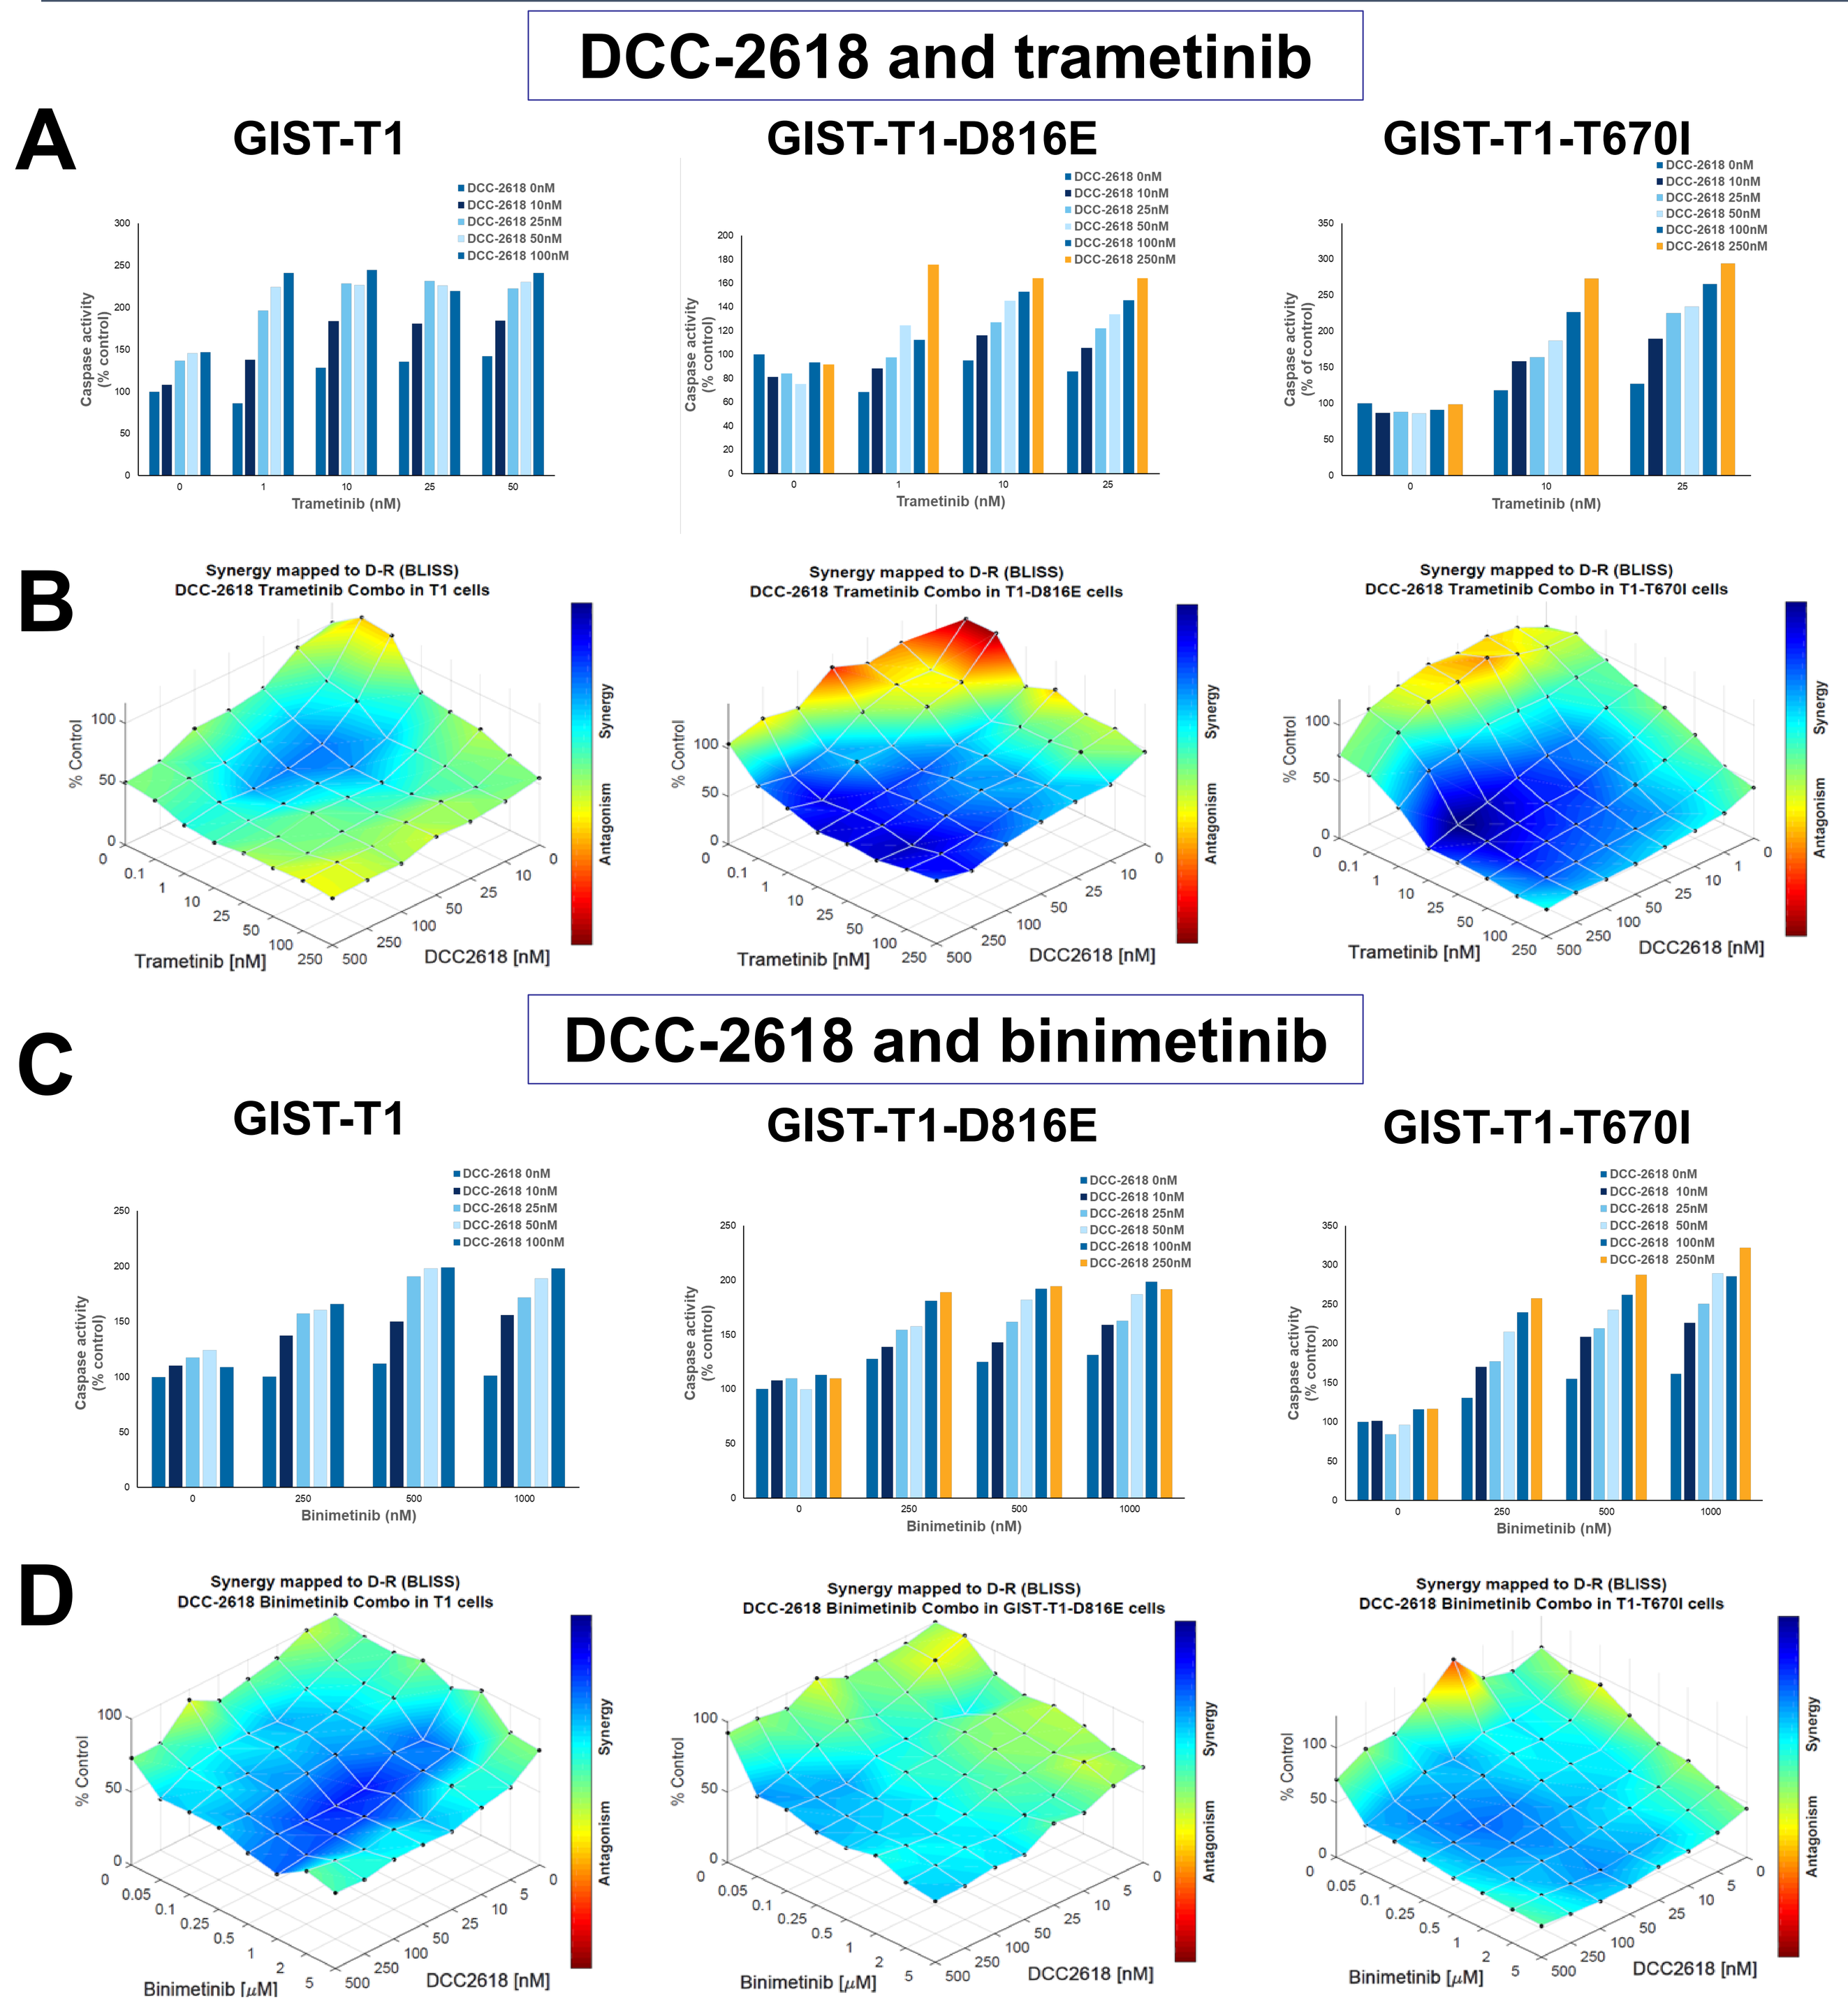
DCC-2618 and trametinib
A
GIST-T1
GIST-T1-D816E
GIST-T1-T670I
Caspase activity (% control)
Trametinib (nM)
B
Synergy mapped to D-R (BLISS)
DCC-2618 Trametinib Combo in T1 cells
DCC-2618 Trametinib Combo in T1-D816E cells
DCC-2618 Trametinib Combo in T1-T670I cells
% Control
Trametinib [nM]
DCC2618 [nM]
Synergy
Antagonism
DCC-2618 and binimetinib
C
GIST-T1
GIST-T1-D816E
GIST-T1-T670I
Binimetinib (nM)
D
DCC-2618 Binimetinib Combo in T1 cells
DCC-2618 Binimetinib Combo in GIST-T1-D816E cells
DCC-2618 Binimetinib Combo in T1-T670I cells
Binimetinib [μM]

Figure 3: DCC-2618 synergizes with MEK inhibitors to induce caspase activity in GIST cell lines. **(A)** Caspase activity in GIST cell lines with DCC-2618 and trametinib combination after 24 h of treatment. Values represent average of three replicates. P values for drug combination vs each drug alone were <0.001. **(B)** Surface mapping of combination dose-responses demonstrate synergy in GIST cell lines with DCC-2618 and trametinib. A matrix of various concentrations of drugs was used to measure caspase activity and the BLISS synergy score was calculated with Combenefit software. **(C)** Caspase 3/7 activity in GIST cell lines with DCC-2618 and binimetinib combination. **(D)** Surface maps of combination treatments to demonstrate synergy.

DCC-2618 synergizes with MEK inhibitors to induce apoptosis in mastocytosis cells

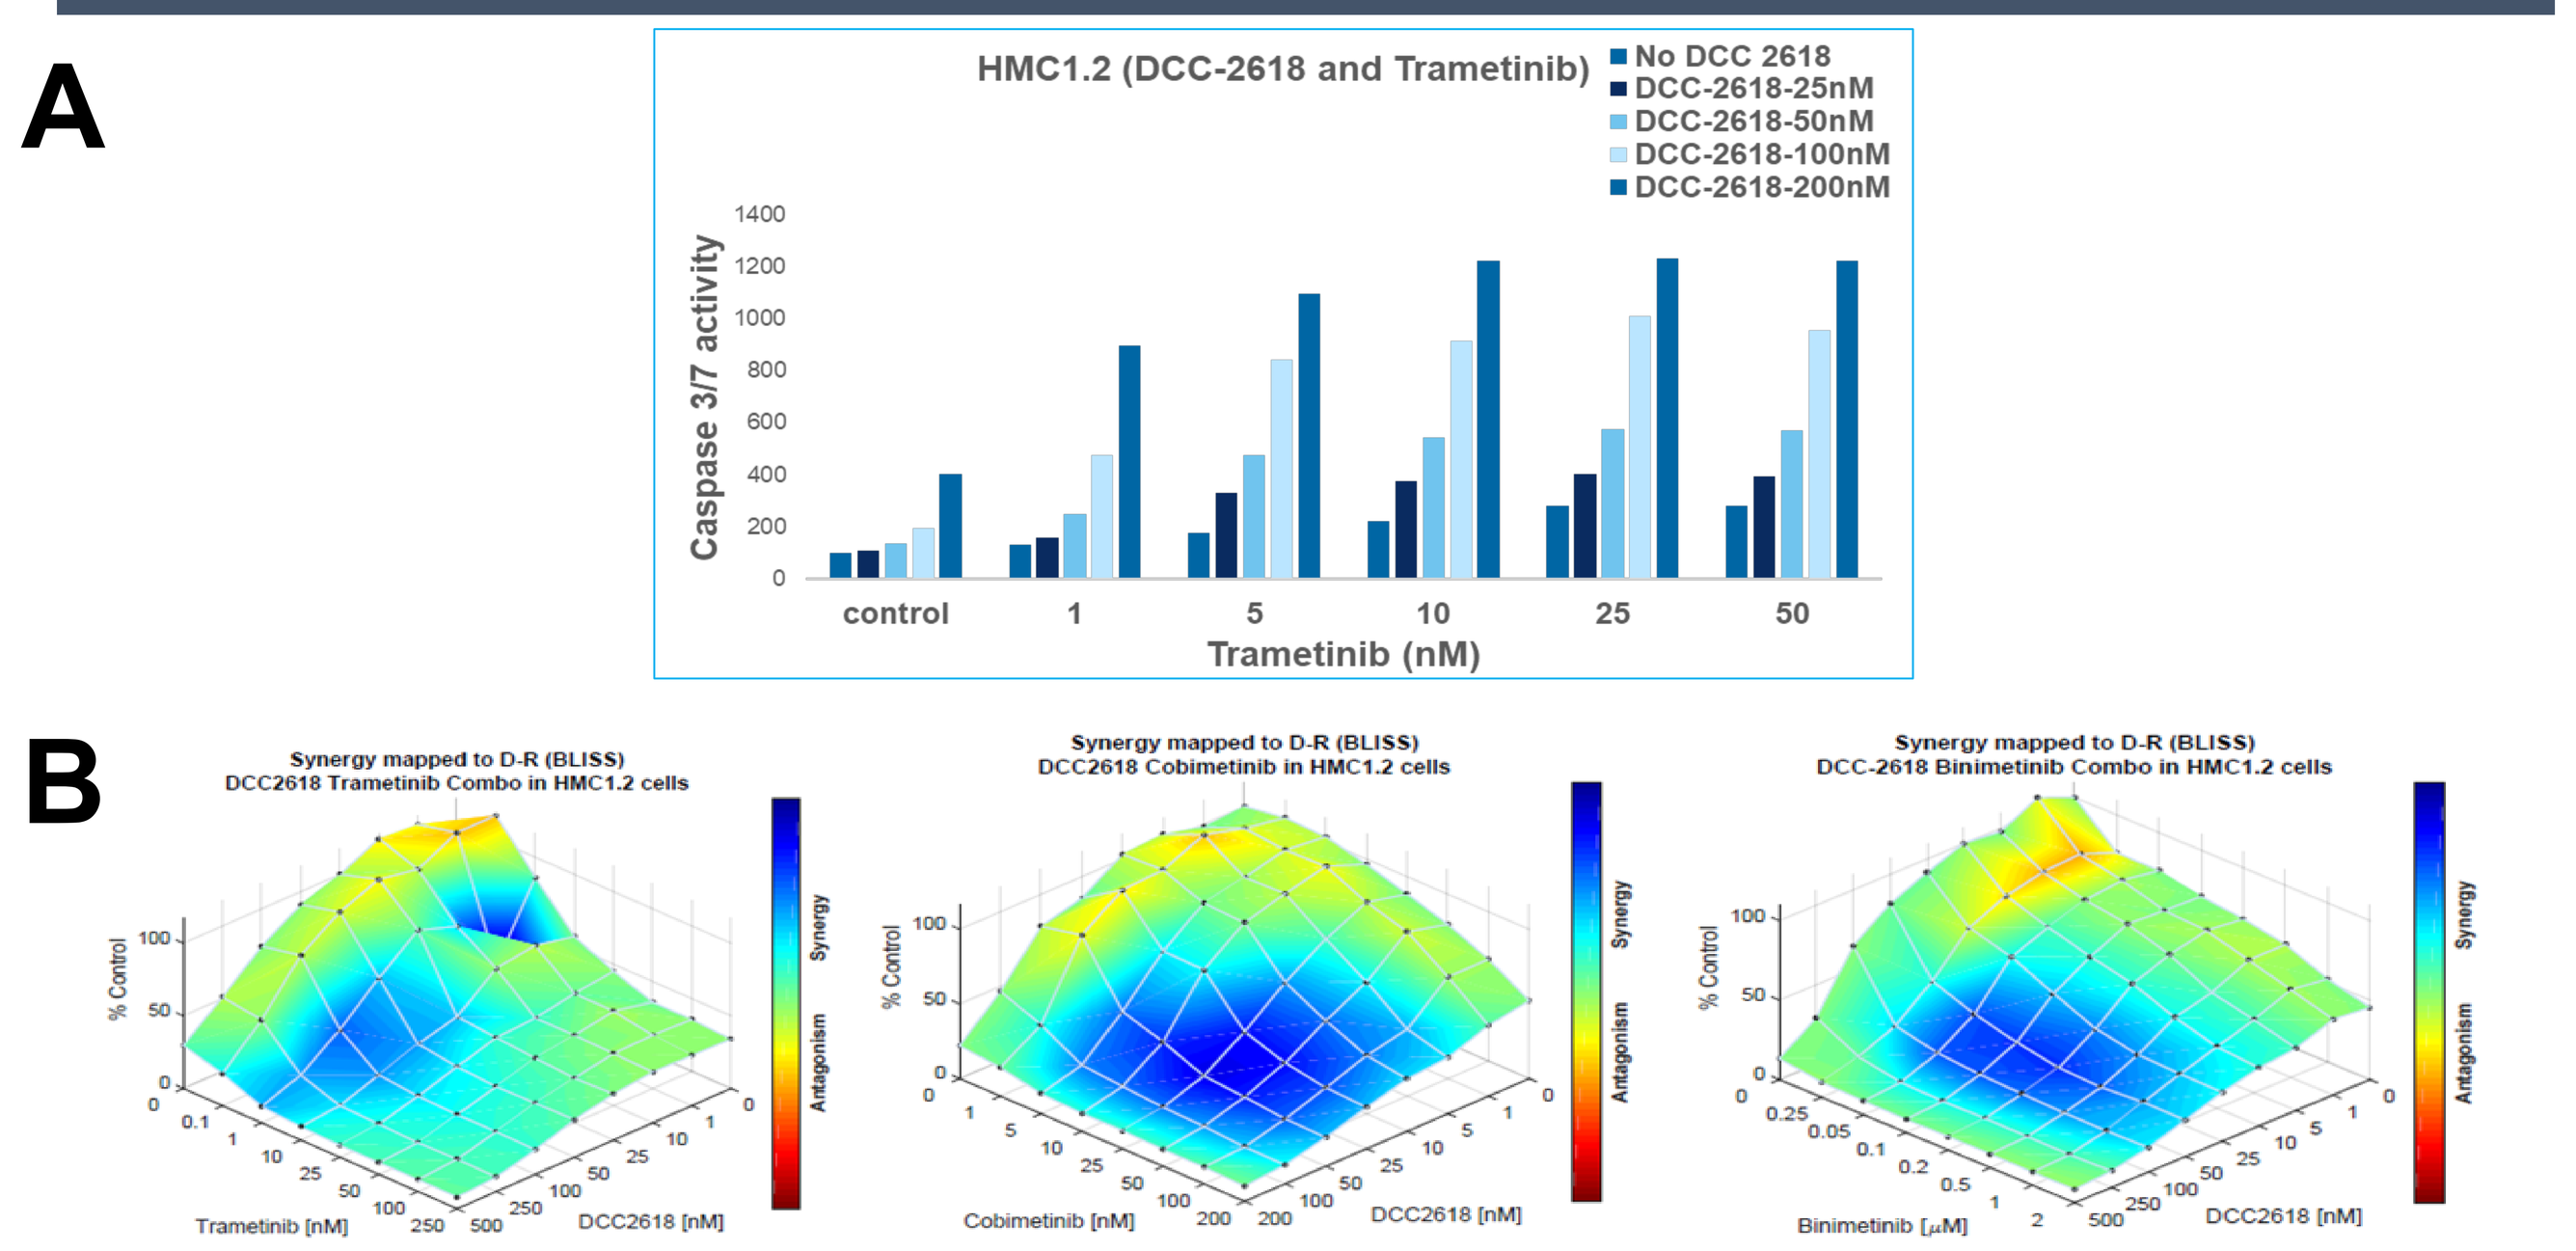
A
HMC1.2 (DCC-2618 and Trametinib)
No DCC 2618
DCC-2618-25nM
DCC-2618-50nM
DCC-2618-100nM
DCC-2618-200nM
Caspase 3/7 activity
control
Trametinib (nM)
B
Synergy mapped to D-R (BLISS)
DCC2618 Trametinib Combo in HMC1.2 cells
DCC2618 Cobimetinib in HMC1.2 cells
DCC-2618 Binimetinib Combo in HMC1.2 cells

Figure 4: DCC-2618 synergizes with MEKi in HMC1.2 cells (V560G/D816V KIT). **(A)** Caspase 3/7 activity in HMC1.2 cells treated with DCC-2618 alone or in combination with trametinib for 24 h. **(B)** Surface map of combination treatments with trametinib, cobimetinib and binimetinib demonstrate synergy.

DCC-2618 in combination with MEK inhibitors prevents colony outgrowth of GIST cells

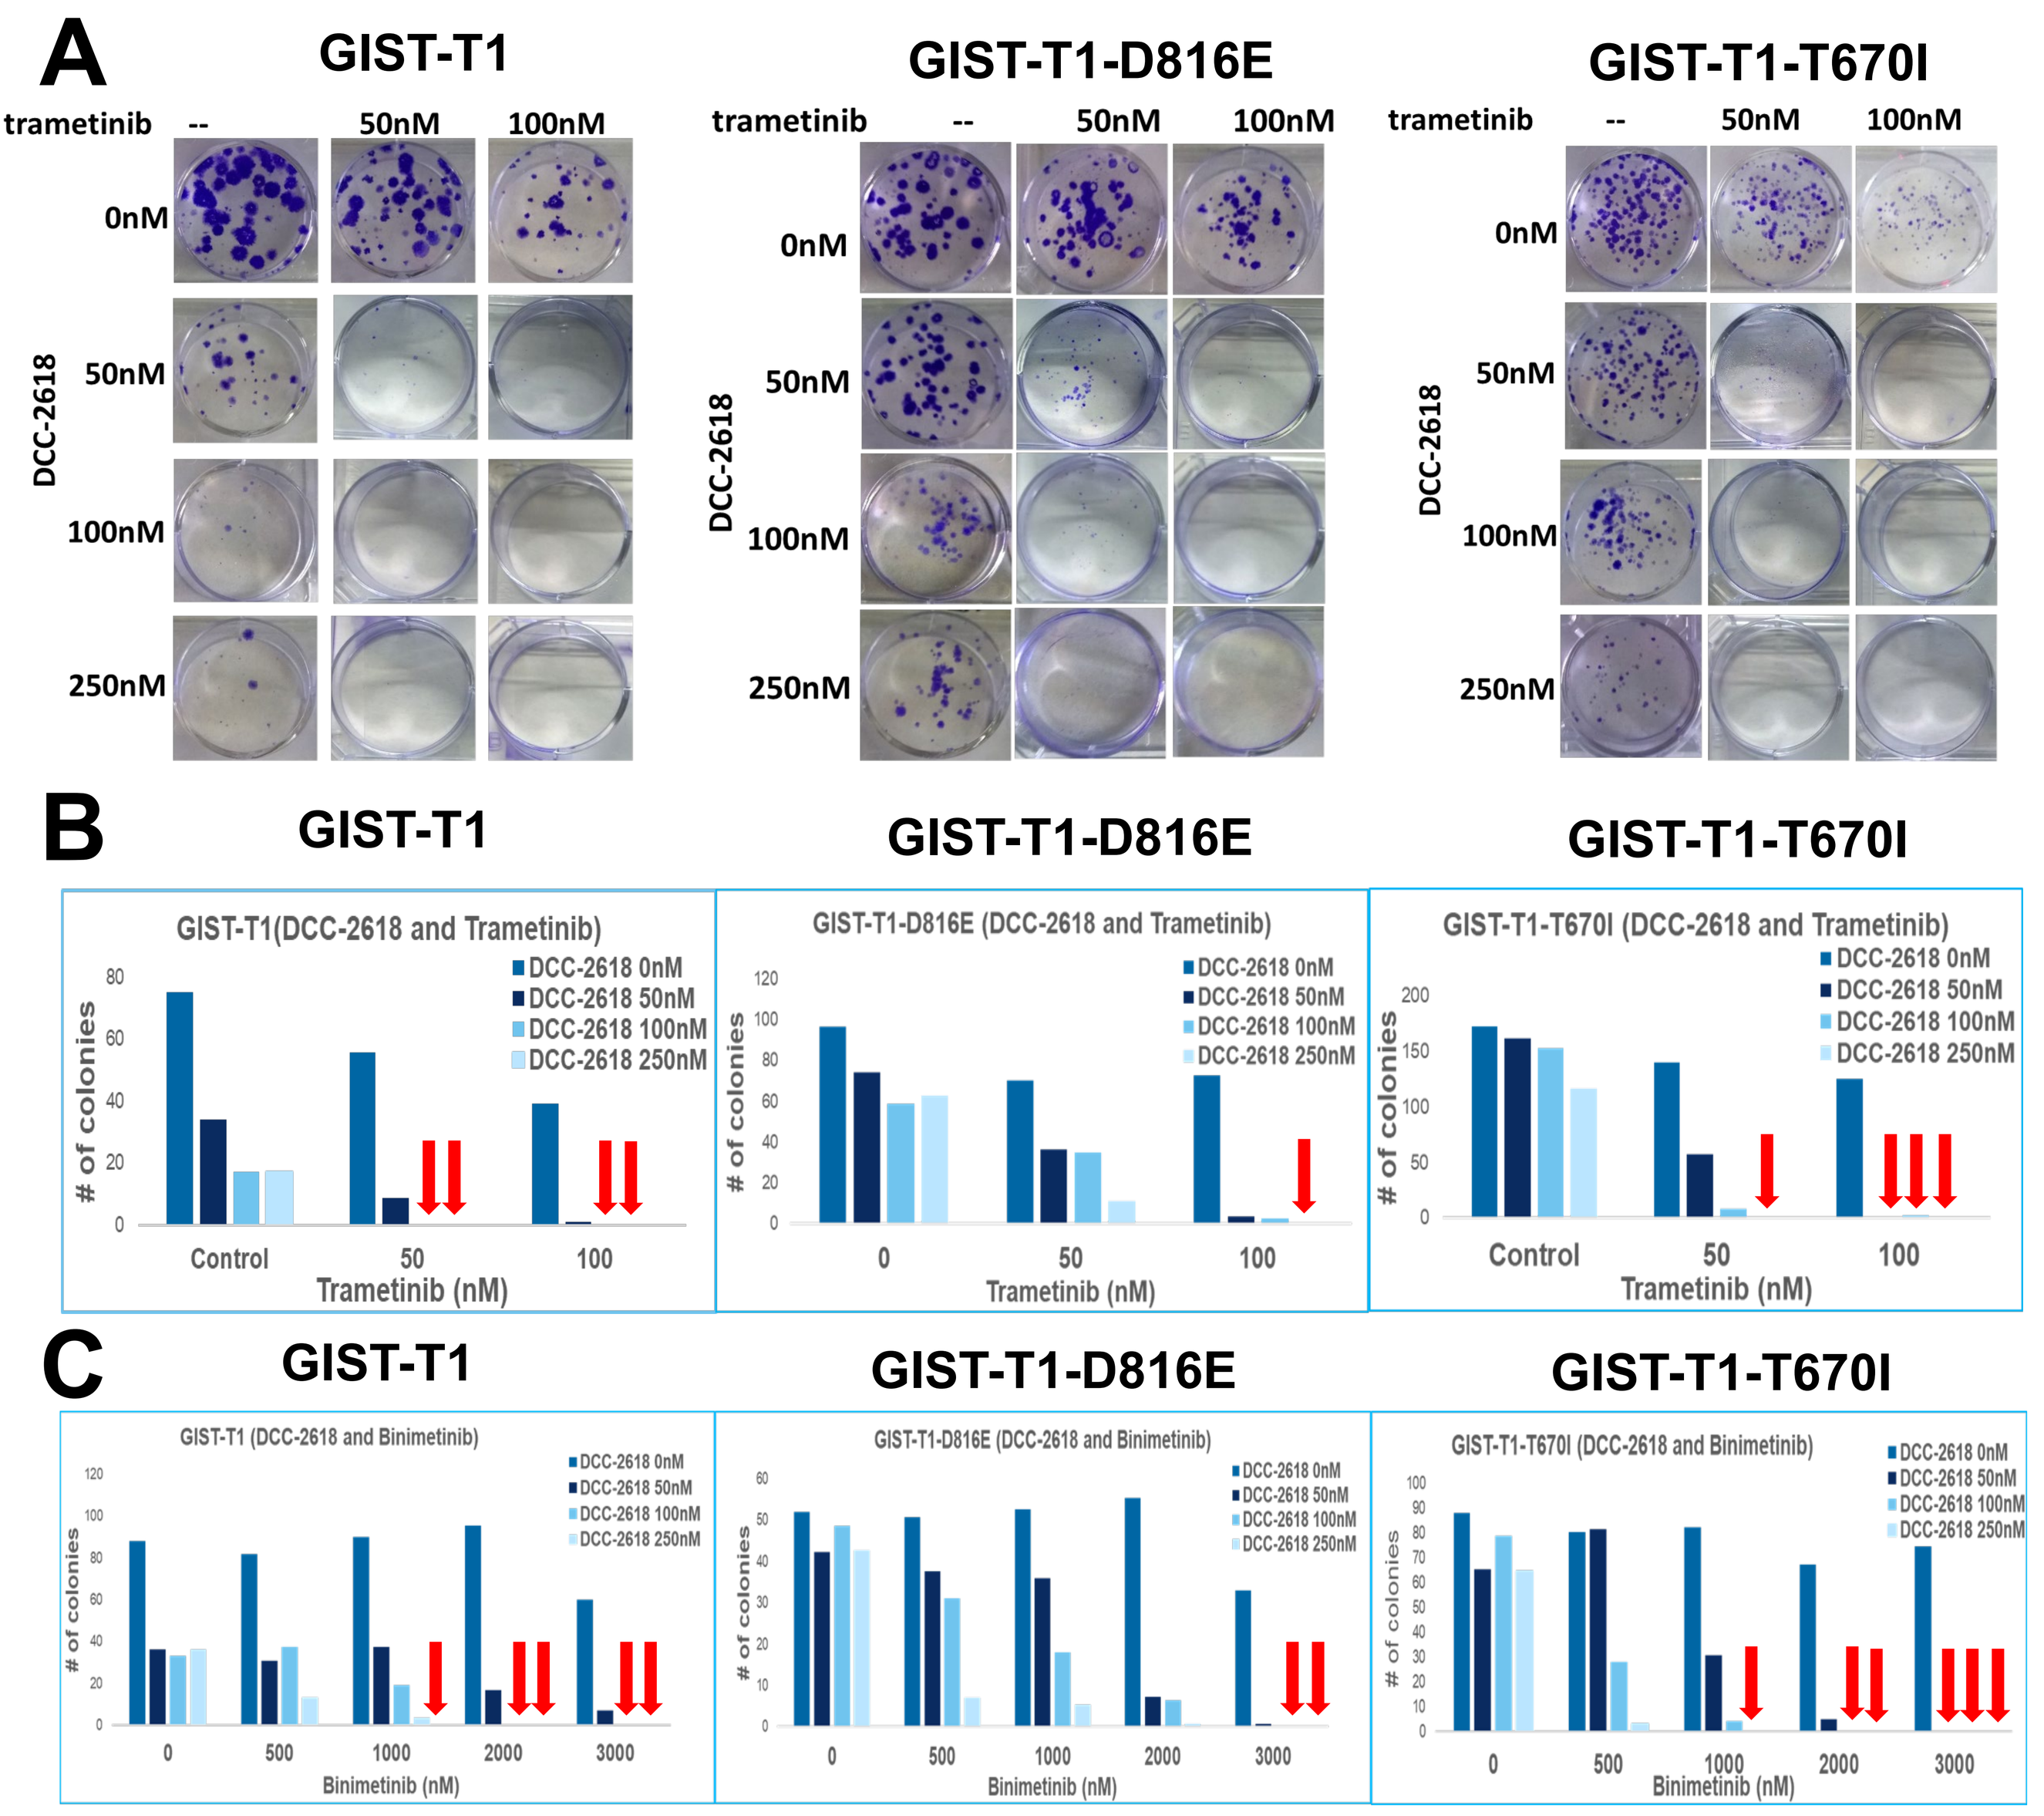
A
GIST-T1
GIST-T1-D816E
GIST-T1-T670I
trametinib
50nM
100nM
DCC-2618
0nM
50nM
100nM
250nM
B
GIST-T1(DCC-2618 and Trametinib)
GIST-T1-D816E (DCC-2618 and Trametinib)
GIST-T1-T670I (DCC-2618 and Trametinib)
of colonies
Trametinib (nM)
C
GIST-T1 (DCC-2618 and Binimetinib)
GIST-T1-D816E (DCC-2618 and Binimetinib)
GIST-T1-T670I (DCC-2618 and Binimetinib)
Binimetinib (nM)

Figure 5: 100 cells for GIST-T1 and GIST-T1-D816E and 500 cells for GIST-T1-T670I were plated in 6 well plates and treated with compounds for 2 weeks. Drugs were washed off and cells were grown in complete media for an additional 10 days. Colonies were stained with crystal violet and counted. **(A)** Colony outgrowth in GIST cell lines treated with DCC-2618 and trametinib. **(B)** Average colony counts from three wells. **(C)** Average colony counts of GIST cell lines treated with a combination of DCC-2618 with binimetinib. Arrows indicate no visually determined colony outgrowth.

DCC-2618/MEKi combination induces apoptosis in N-Ras G12D expressing mastocytosis cells

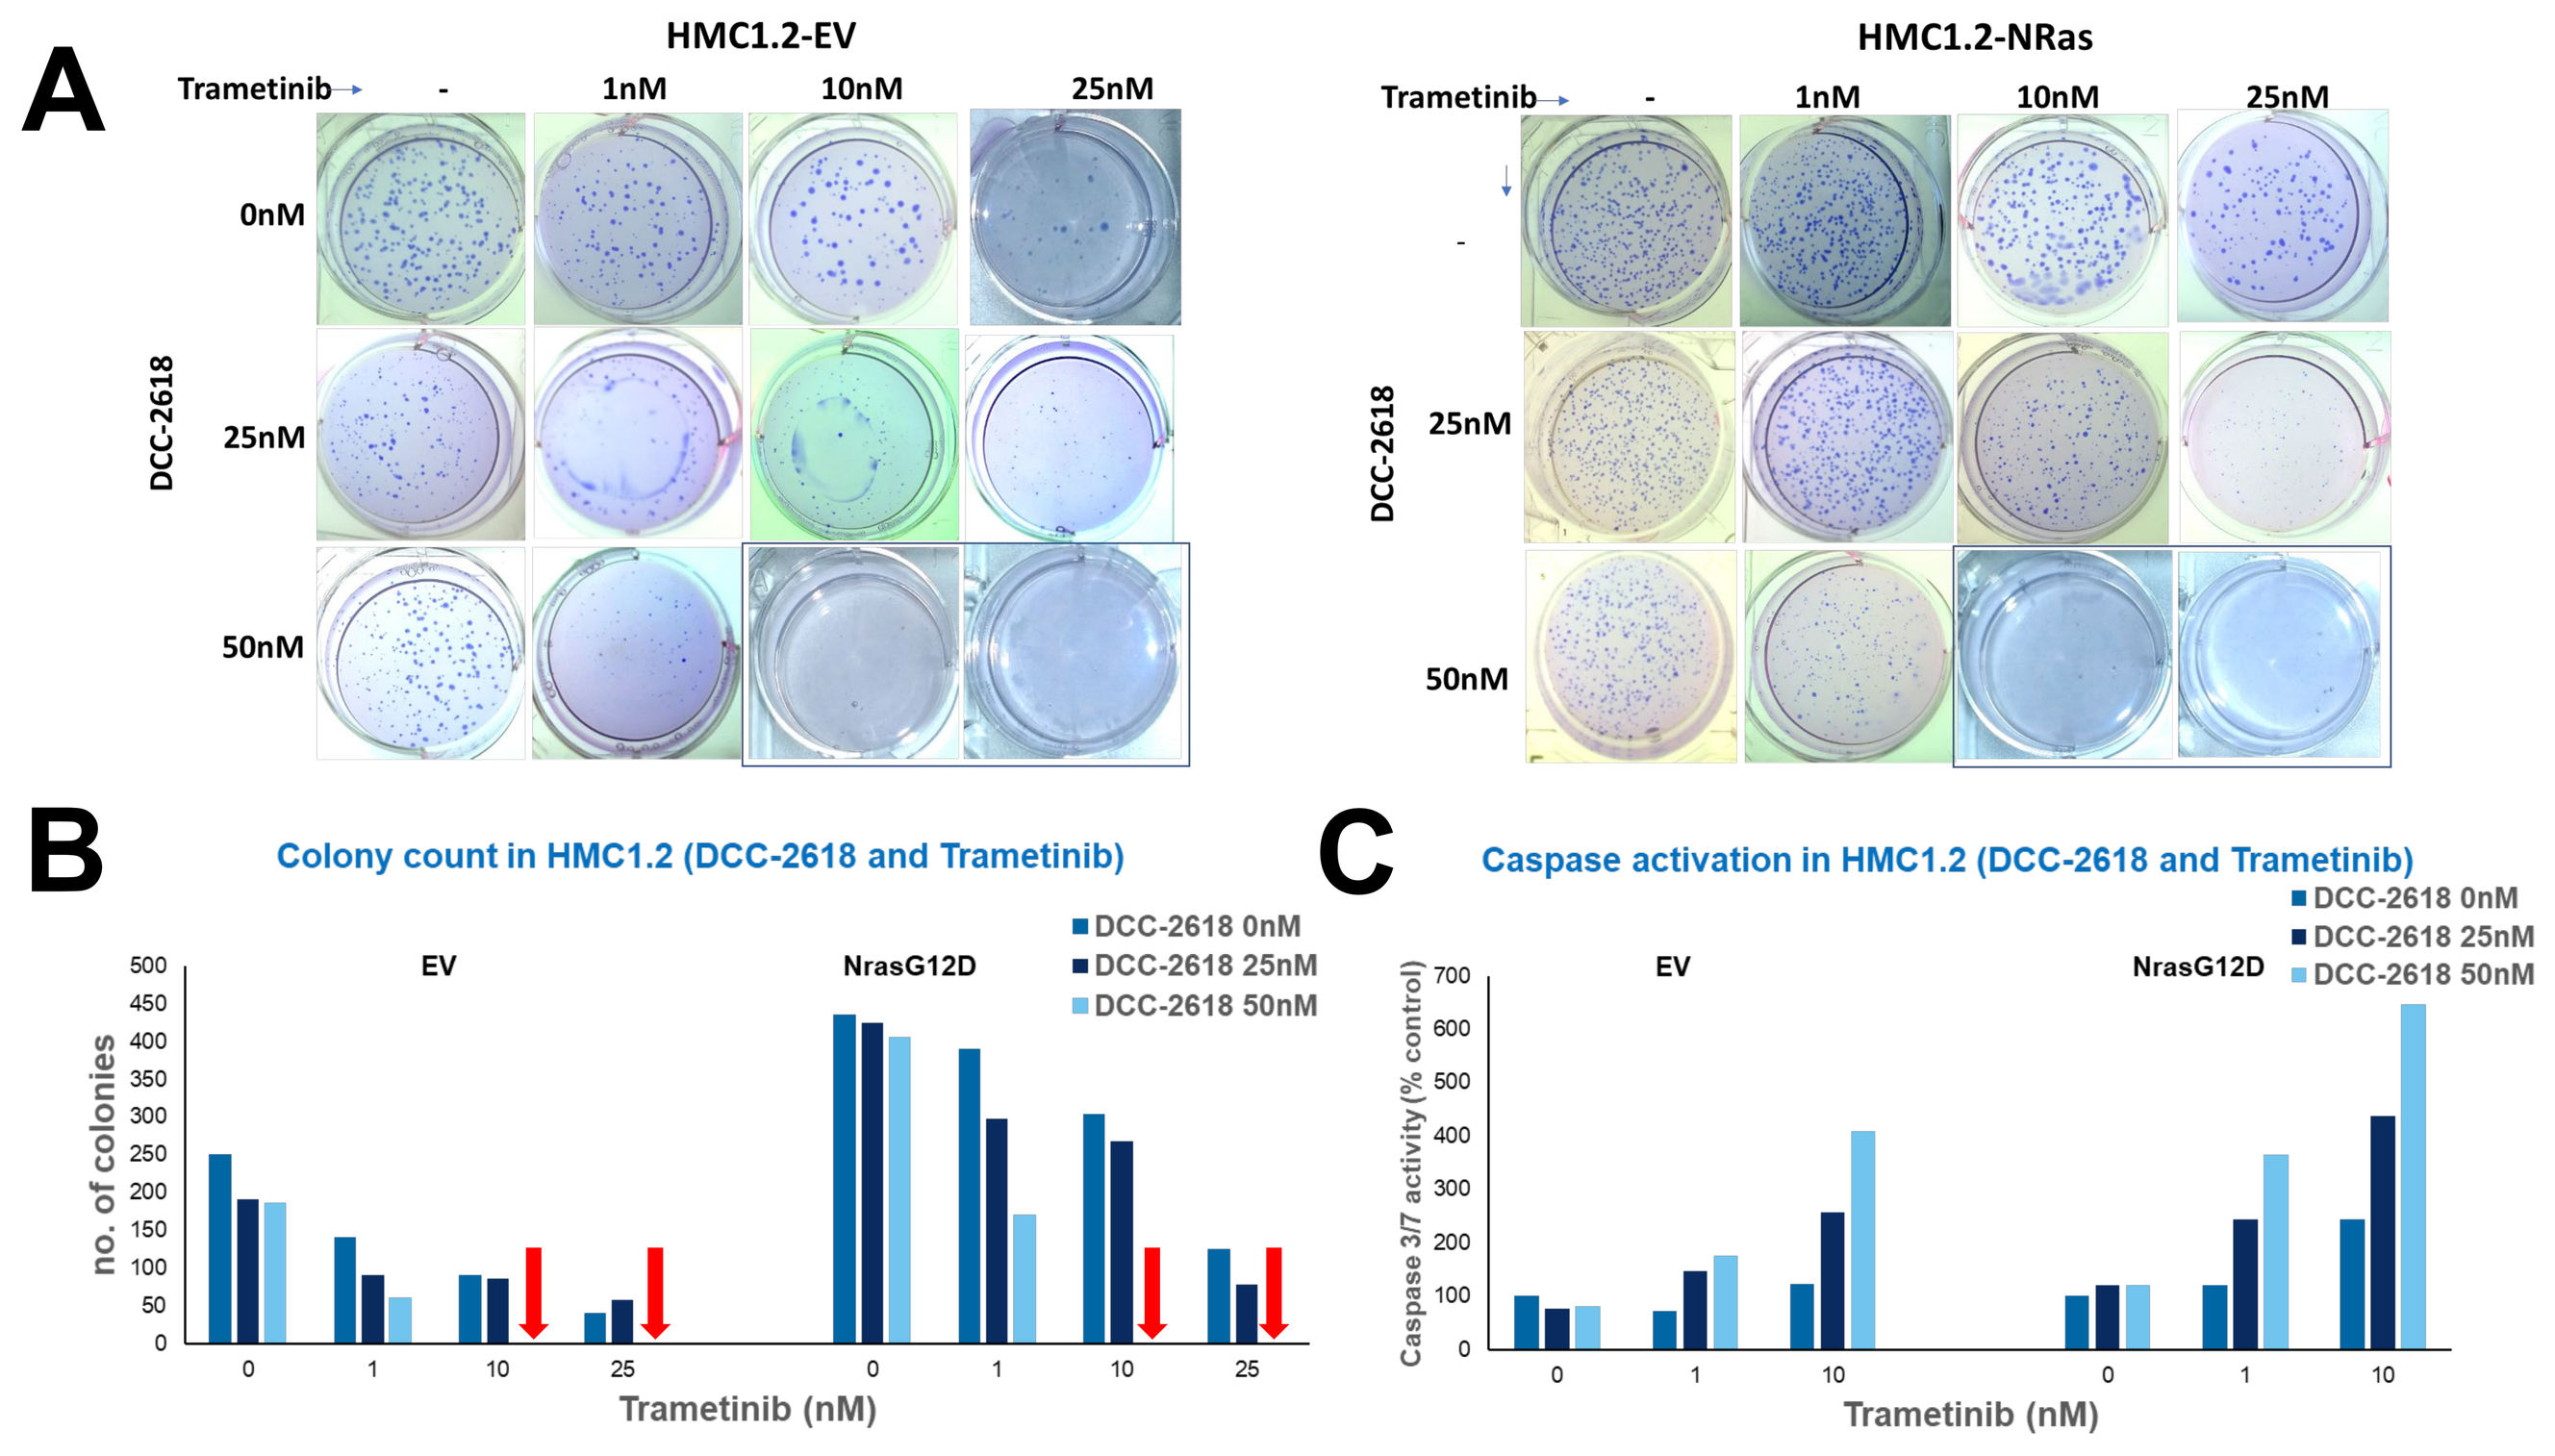
A
HMC1.2-EV
HMC1.2-NRas
Trametinib
1nM
10nM
25nM
DCC-2618
0nM
25nM
50nM
B
Colony count in HMC1.2 (DCC-2618 and Trametinib)
EV
NrasG12D
no. of colonies
Trametinib (nM)
C
Caspase activation in HMC1.2 (DCC-2618 and Trametinib)
Caspase 3/7 activity (% control)

Figure 6: Combination treatment inhibits colony outgrowth in N-Ras G12D transfected HMC1.2 cells. **(A)** HMC1.2 empty vector (EV) control and N_ras mutant G12D transfected cells were grown in soft agar for 10 days in the presence of DCC-2618 and trametinib. Drug was washed off and cells were grown for 5 days in complete media. Colonies were stained and photographed. **(B)** Average colony counts from 3 wells. **(C)** Caspase activity in EV and N-Ras G12D transfected cells in combination with DCC-2618 and trametinib.

Saturation Mutagenesis

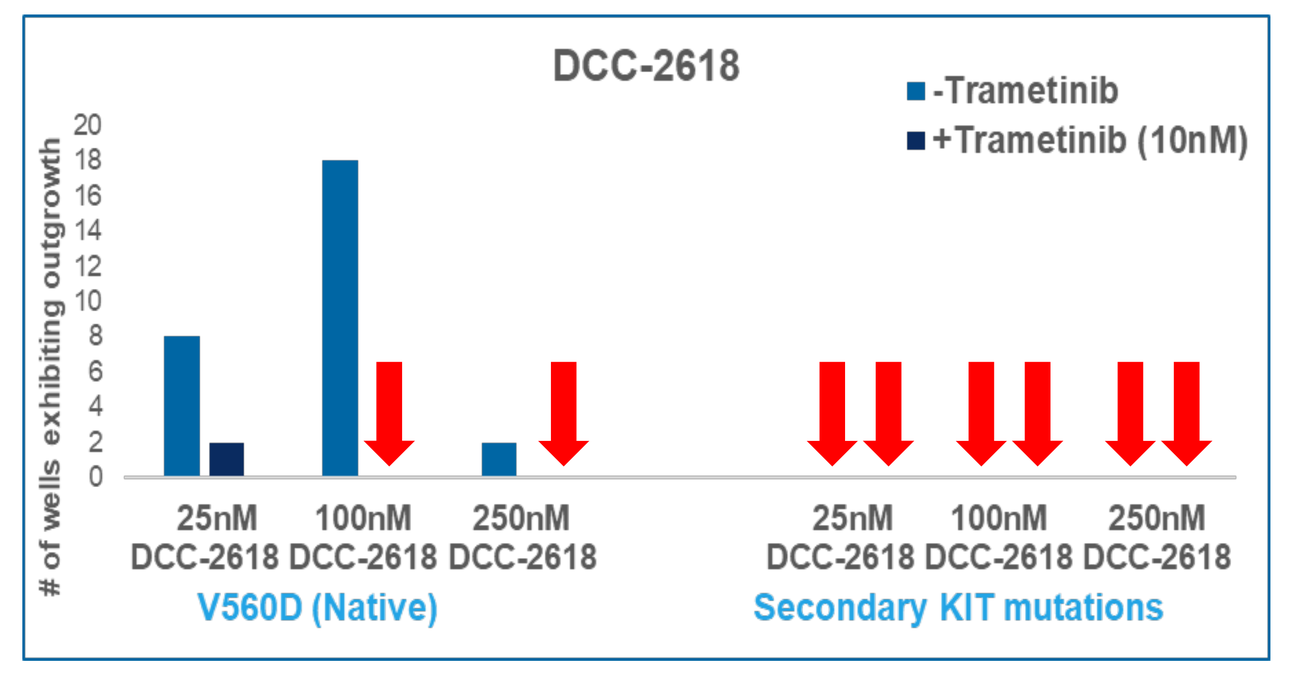
DCC-2618
-Trametinib
+Trametinib (10nM)
of wells exhibiting outgrowth
25nM DCC-2618
100nM DCC-2618
250nM DCC-2618
V560D (Native)
Secondary KIT mutations

Figure 7. Combination of DCC-2618 and trametinib prevents emergence of drug resistance mediated by non-KIT mechanisms. Ba/F3 cells stably expressing V560D KIT were treated with ENU and grown in the presence of DCC-2618 +/- trametinib for 28 days. Wells exhibiting outgrowth were sequenced for KIT mutations. "V560D (Native)" indicates that no secondary mutations were found within the KIT gene, and mutations may have arisen in other pathways, such as the MAPK pathway, that allow for Ba/F3 cell growth. Arrows indicate no outgrowth.

DCC-2618 in combination with trametinib inhibits GIST xenograft tumor growth

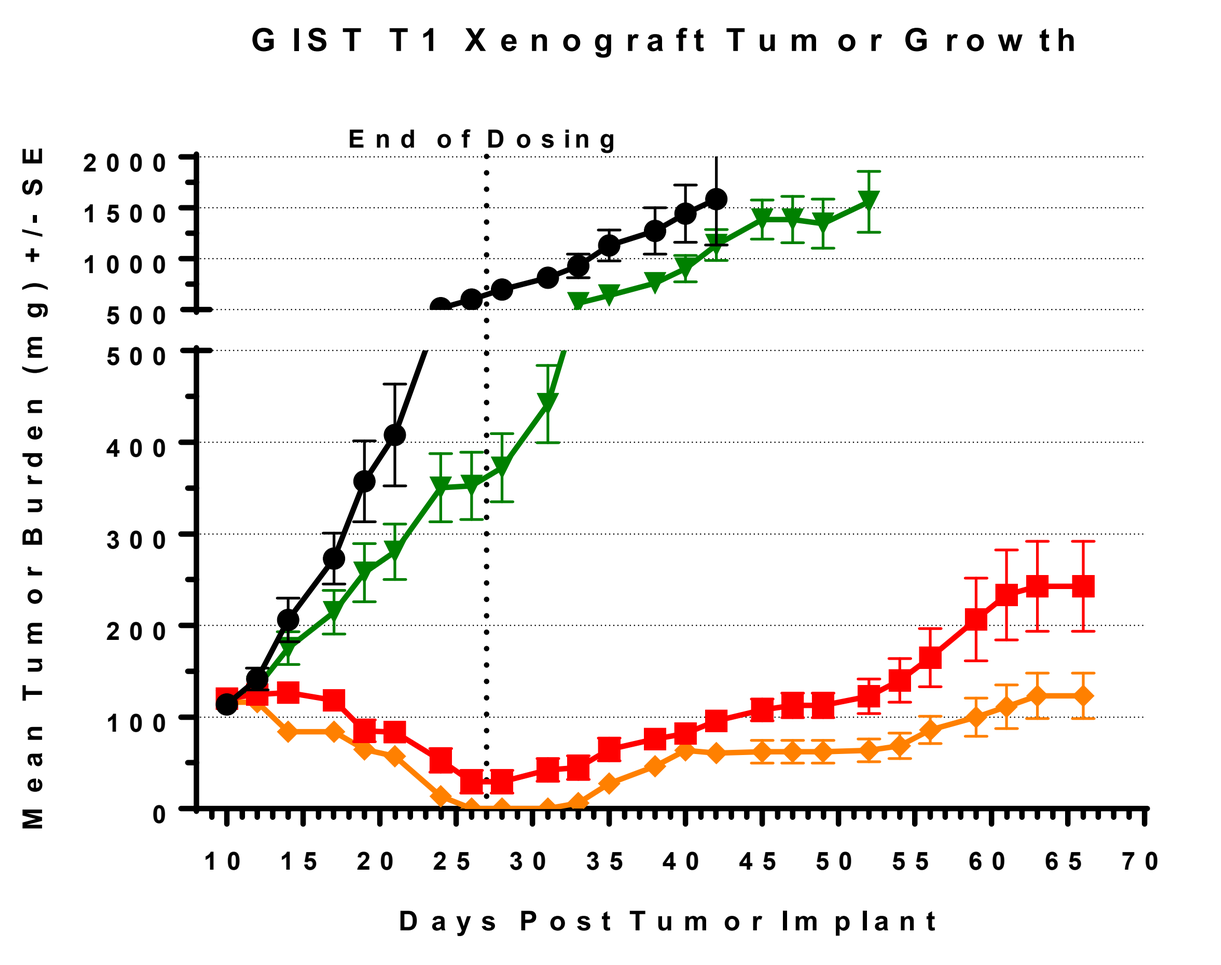
GIST T1 Xenograft Tumor Growth
End of Dosing
Mean Tumor Burden (mg) +/- SE
Days Post Tumor Implant
Vehicle
DCC-2618 ~100 mg/kg chow
trametinib 0.5 mg/kg PO BID
DCC-2618 ~100 mg/kg + trametinib

	Day 27		Day 67	
	PR	CR	PR	CR
DCC-2618	4	6	2	0
Trametinib	0	0	0	0
DCC-2618 + trametinib	0	10	4	1

Figure 8. DCC-2618 and trametinib treatment results in tumor regressions in a GIST-T1 xenograft model. Mice were treated with DCC-2618 formulated into the diet to achieve ~100 mg/kg/day (red), trametinib at 0.5 mg/kg BID PO (green), or with the combination of drugs (orange). Dosing started on day 10 and ended on day 27. Tumor growth was monitored for additional 40 days. Partial and complete tumor regressions (PR and CR) are shown at Days 27 and 67.

Summary

- KIT inhibitors are cytostatic in GIST cells, and cells start to grow back when drug is removed. The combination of DCC-2618 with a MEK inhibitor induces apoptosis and prevents the outgrowth of colonies even after drug is removed.
- DCC-2618 shows strong synergy in inducing apoptosis in combination with MEK inhibitors trametinib and binimetinib in imatinib-sensitive and, importantly, in imatinib-resistant GIST and mastocytosis cell lines.
- Ras is frequently mutated in advanced forms of systemic mastocytosis and Ras mutant expressing HMC1.2 cells were more sensitive to the combination of DCC-2618 and trametinib.
- In saturation mutagenesis studies, treatment with DCC-2618 prevents the outgrowth of cells with drug-resistant KIT mutations. A few wells did exhibit outgrowth, in which cells presumably had mutations in other signaling proteins or pathways, such as the MAPK pathway. The combination of DCC-2618 with trametinib completely suppressed clonal growth.
- The combination of DCC-2618 and trametinib in a GIST-T1 xenograft model leads to more partial and complete tumor regressions than single agent treatment, even with a short treatment period of 18 days and 40 days of allowing tumors to regrow after dosing was stopped.

Contact bsmith@deciphera.com for permission to reprint and/or distribute.