Translational research in a phase I proof-of-concept study supports that DCC-2618 is a pan-KIT inhibitor

Filip Janku[1], Albi Razak[2], Michael Gordon[3], David Brooks[4], Daniel Flynn[5], Anu Gupta[5], Michael Kaufman[5], Cynthia Leary[5], Bryan Smith[5], Deb Westwood[4], Neeta Somaiah[6], Elena Helman[7], Oliver Rosen[4], Suzanne George[8]

[1]The University of Texas MD Anderson Cancer Center, Department of Investigational Cancer Therapeutics, Houston, TX, USA, [2]Princess Margaret Cancer Centre, Cancer Clinical Research Unit, Toronto, Canada; [3]Pinnacle Oncology Hematology, Phase 1 Unit, Scottsdale, AZ, USA; [4]Deciphera Pharmaceuticals, Clinical Research & Development, Waltham, MA, USA; [5]Deciphera Pharmaceuticals, Research, Lawrence, KS, USA; [6]The University of Texas MD Anderson Cancer Center, Department of Sarcoma Medical Oncology, Houston, TX, USA; [7]Guardant Health, Redwood City, CA, USA; [8]Dana-Farber Cancer Institute, Center for Sarcoma and Bone Oncology, Boston, MA, USA

Abstract #LB-039

BACKGROUND

- DCC-2618 is a KIT and PDGFRA inhibitor resilient to de-novo and drug resistance mutations and independent of ATP concentration.
- DCC-2618 is a switch control inhibitor, designed to potently inhibit a broad range of mutations in KIT and PDGFR kinases, to exhibit long inhibitory residence times.
- The active metabolite of DCC-2618, DP-5439, is a major contributor to total human drug exposure and exhibits comparable activity across all KIT mutations.
- Gastrointestinal stromal tumor (GIST) is an important disease to achieve proof-of-concept due to the multiplicity and heterogeneity of resistance mutations within *KIT.*
- In heavily pretreated GIST patients, DCC-2618 produced a high rate of Partial Metabolic Response (PMR) by FDG-PET and disease control in GIST refractory to TKIs[1]. We provide further evidence of the diversity of drug resistant KIT mutants and the heterogeneity within individual patients and measure the effect of treatment on cell-free (cf) DNA allele frequency of mutant *KIT* and *PDGFR* and on the prevalence of circulating tumor cells (CTC).
- The C_{min} of total exposure to DCC-2618 and metabolite is correlated with changes in cfDNA *KIT* mutant allele frequencies (MAF) compared to baseline values across the spectrum of exons 9, 11, 13, 14, 17 and 18 starting at the 30 mg twice daily (BID) cohort.

METHODS

- ✓ Plasma cfDNA: Mutations were detected and quantitated by Guardant 360 v2.9 or v2.10. This 73 gene panel was analyzed by next generation sequencing. Only amino acid-altering mutations in *KIT* and *PDGFRA* are reported here. Changes in the frequency of *KIT* or *PDGFRA* mutations are reported from baseline levels except where noted.
- ✓ CTC: Whole blood was enriched for CTCs in an OncoQuick tube. The CTC layer was incubated with an adenovirus that replicates and expresses GFP in cells with high levels of telomerase (Oncolys BioPharma Inc.). Cells were then incubated with fluorescently-labeled antibodies, fixed, and stained with DAPI. CTCs positive for DAPI, GFP, KIT, PDGFRA, or DOG1 fluorescence were counted using a BioTek Cytation 5 imager.

RESULTS

Figure 1: Translation of 'bench' results to the bedside: Predicted potency of DCC-2618 against selected *KIT* secondary mutations (right) compared to exposure observed in lower cohorts of FIH study (left) and reduction of mutation allele frequency (MAF) of selected mutations in FIH study (center)

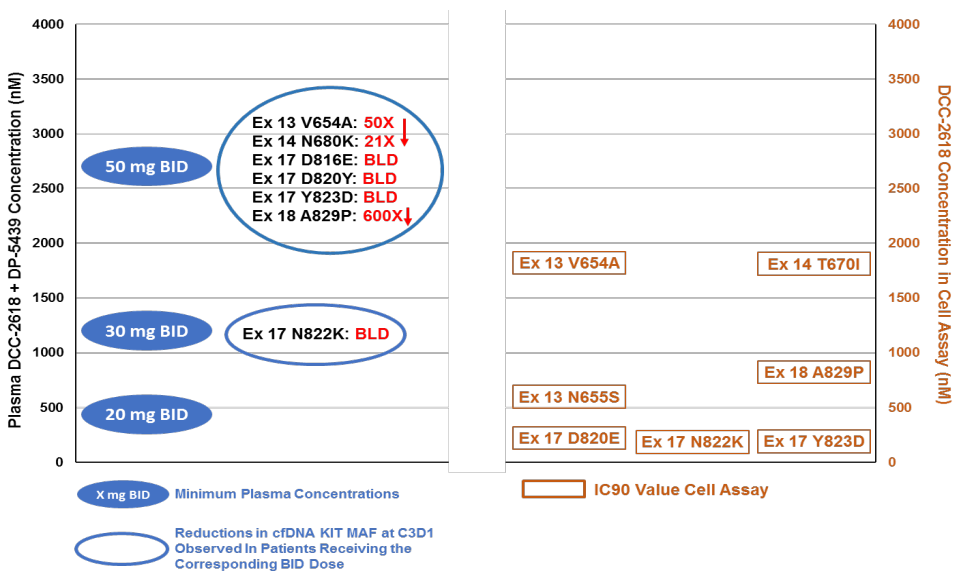
Plasma DCC-2618 + DP-5439 Concentration (nM)
DCC-2618 Concentration in Cell Assay (nM)
50 mg BID
30 mg BID
20 mg BID
Ex 13 V654A: 50X
Ex 14 N680K: 21X
Ex 17 D816E: BLD
Ex 17 D820Y: BLD
Ex 17 Y823D: BLD
Ex 18 A829P: 600X
Ex 17 N822K: BLD
Ex 13 V654A
Ex 14 T670I
Ex 18 A829P
Ex 13 N655S
Ex 17 D820E
Ex 17 N822K
Ex 17 Y823D
X mg BID Minimum Plasma Concentrations
Reductions in cfDNA KIT MAF at C3D1 Observed In Patients Receiving the Corresponding BID Dose
IC90 Value Cell Assay

C_{min} plasma concentrations were determined on C1D15 and are limited to cohorts from 20 to 50 mg BID. The IC90s of KIT secondary mutations were obtained in transfected CHO cells. Reduction of MAF was determined as described under 'Background.' BLD = Below Level of Detection.

Figure 2: Example of GIST CTC: KIT-mutant patient – Positive for both KIT and GIST Marker DOG1

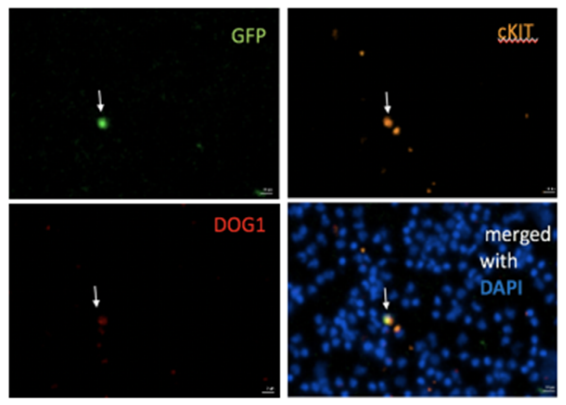
GFP
cKIT
DOG1
merged with DAPI

Detection of CTCs positive for GFP (upper left), KIT (upper right), or DOG1 (lower left) and merged fluorescence images with DAPI stain (lower right).

Reference: [1]EORTC-NCI-AACR Symposium 2016: 7LBA

Table 1: Comparison of cfDNA and CTC results with PET scans, CT scans, and plasma concentrations

			PET Scan	CT Scan				cfDNA Mutant Allele Fraction				CTCs/mL	CTC Levels (change from baseline)			
ID / Gene	Dose (mg BID)	Mutated Exons	C1D21	C3D1	C5D1	C7D1	EOS	C3D1	C5D1	C7D1	EOS	Baseline	C3D1	C5D1	C7D1	EOS
3 / KIT	20	11, 13	ND	PD				↑2-3X				Not done				
7 / KIT	30	11, 17	PMR	SD	SD	PD		↓3-16X	↑7-16X	↓3-16X	NR	2.3	↓59%	↑65%	↑91%	
9 / KIT	50	9, 17	PMR	SD	SD	-	-	↓16-21X				10.2	↓84%			
12 / KIT	50	11	PMR	SD	SD	SD	PD	↓3X	↓3X		↑12X	5.2	-	↓52%		↑36%
11 / KIT	50	11, 13, 18	PMR	SD	SD	SD		↓50->600X	↓136->600X	↑7-16X		14.1	↑56%	↓100%	↓95%	
10 / KIT	50	11 (2x), 17 (4x)	PMR	SD	-	-	-	↓1-20X				3.9	↓10%			
13 / KIT	100	None	PMR	PR	PR	PR						2.4	↓23%		↑38%	
14 / KIT	100	9, 17 (2x)	PMR	SD	SD	SD		↓17X				4.6	↓90%	↓61%	↓39%	
18/ PDGFRA	100	18	NA	SD	SD	SD		D842V only at C3D1*	↓6X*	↓6X*		2.5	↓100%	↓100%		
15 / KIT	100	9, 14	PMR	SD	PD	-	PD	↓2-3X				0.8				↑1000%
20 / KIT	150	9	PMR	PD	-	-	PD	↑9X			↑9X	80				↑2270%
22 / KIT	150	9	PMR	SD	SD			Not done				6.2		↓91%		
23 / KIT	150	18	PMR	SD	PR	PR		↓7X	↓7X			6.3		↓72%	↓100%	
19 / KIT	150	None	PMR	SD	SD			Not done				8.2		↓45%		
21 / KIT	150	11, 13, 14, 17 (3x)	PMD	SD	SD			≥ 3X↓				12.4		↓99%		
24 / KIT	150	11, 17 (2x)	PMR	-	SD			Not done				201		↓95%		

Plasma cfDNA revealed a total of 43 *KIT* mutations in 20 of 24 (83%) patients with KIT mutant GIST (not all shown). *KIT* mutations from cfDNA included previously documented initiating mutations in exons 9, 11 as well as resistance mutations in exons 13, 14, 17, and 18. The range was from 0-6 *KIT* mutations (see Mutated Exons column above) with mean ~1.9 *KIT* mutations per patient with cfDNA. DCC-2618 treated patients had a decline in *KIT* or *PDGFRA* D842V mutant allele fraction in 9 of 10 patients with samples from more than one time point. In 6 of these 8 patients with declines at C3D1 (the first time point examined on study), at least one *KIT* mutant allele fraction fell below the limit of detection of the assay (0.1% allele fraction) indicating rapid clearance of mutations from plasma. The single patient (#20) with increase in plasma *KIT* mutation (9x at C3D1) was coincident with Progressive Disease on CT scan and a sharp rise in KIT+ CTCs all at C3D1.

* D842V was detected in plasma from patient 18 with PDGFRA mutant GIST for the first time at C3D1 and subsequently cleared.

Figure 3: 'Niagara Fall' plot for KIT Mutations

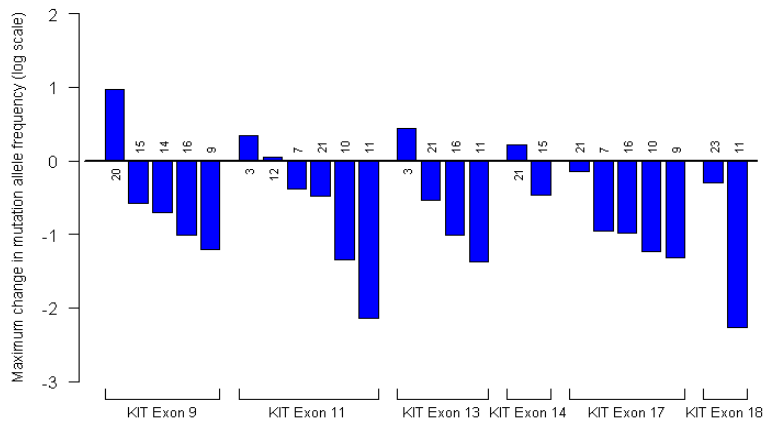
Maximum change in mutation allele frequency (log scale)
KIT Exon 9
KIT Exon 11
KIT Exon 13
KIT Exon 14
KIT Exon 17
KIT Exon 18

Maximum change of cfDNA mutation allele frequency for each *KIT* exon

Note: Patient IDs are presented with each bar (See Table 1 for patient data).

Figure 4: Correlation of tumor volume with KIT MAF and CTCs in a single patient

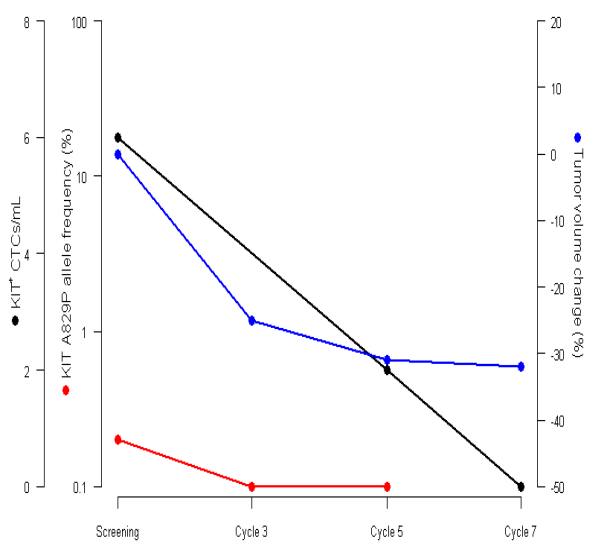
KIT+ CTCs/mL
KIT A829P allele frequency (%)
Tumor volume change (%)
Screening
Cycle 3
Cycle 5
Cycle 7

Tumor volume change according to RECIST per investigator assessment (blue) is compared to change in KIT A829P MAF (red) and change in CTCs (black) in patient 23 of Table 1 (change from baseline).

CONCLUSIONS

- cfDNA might have the potential for monitoring of treatment outcome. Currently, available data are not yet mature to determine whether it can be used to guide treatment decisions.
- Currently available TKIs primarily inhibit either the ATP binding pocket mutations (exon 13/14) or the activation loop (exon 17/18) and do not demonstrate activity across both regions which are known to cause imatinib resistance in GIST thereby leaving significant liabilities in inhibitory coverage of known KIT resistance mutations.
- In patients with heavily pre-treated TKI resistant GIST, DCC-2618 has led to notable decreases in MAF of resistance mutations in both the activation loop (exon 17/18) and ATP-binding pocket mutations (exon 13/14) supporting the role of DCC-2618 beyond imatinib resistance.
- The V654A KIT mutation is in vitro among the least sensitive KIT mutant to DCC-2618 (cellular IC50 189 nM). Clearance of cfDNA V654A MAF demonstrates high Cmin at 50 mg BID sufficient to treat V654A positive GIST lesions (Figure 1).
- Our data provide a first evidence of GIST CTC based on KIT/PDGFRA and DOG1 positivity.

Acknowledgment: We would like to thank the patients, their families, and the site staff of the DCC-2618-01-001 trial.